CELx™ | breast
P13K Signaling Function Test

Sub-Group of PIK3CA WT breast cancer patients have hyperactive S1P and LPA signaling tumors responsive to PI3K inhibitors: functional signaling test identifies new patient group who may benefit from PI3K inhibitors

Lance G Laing, Salmaan Khan, Ian A MacNeil, Catherine Kuzmicki, Samantha Kharbush, Benjamin Rich, Abhay Shukla, Kelly Brass, Brian Sullivan | Celcuity, 16305 36th Ave N, Suite 100, Minneapolis, MN 55446, USA

celcuity™
www.celcuity.com

Background

Less than 20% of PI3KCA mutated late stage breast cancer patients achieved an objective response in a Phase III clinical trial with alpelisib, a recently approved PIK3CA inhibitor.[1] This suggests factors other than PIK3CA sequence variance status may be important to measure when identifying patients eligible for PI3K inhibitors. The work described here examines aberrant GPCR-linked signaling through PI3K isoforms for potential identification of patients with PI3K-linked disease activity.

Class I PI3K isoforms have been shown to cooperate with GPCRs in several different ways to regulate signaling and oncogenic transformation. In one significant report, PI3K p110γ and p110β have been shown to be activated directly by GPCR signaling. Residues in p110γ and its regulatory isoform, p101, have been shown to be critical for direct interaction with GPCR Gβγ heterodimer driven signaling and oncogenic transformation.[2] Additionally, PI3K activity in epithelial cells is directly linked to adhesion and concomitant cytoskeletal rearrangements.[3]

The accumulation of phospholipid agonists of specific GPCRs, due to lipid metabolism abnormalities in cancer cells, may be a causal factor in tumor progression.[4] Sphingosine 1-phosphate (S1P) and Lysophosphatidic Acid (LPA) are bioactive lipid agonists that signal through binding to two important GPCR families, S1PR 1-5 and LPAR 1-6, respectively. S1P and LPA are able to signal through their specific GPCR receptors in a paracrine or autocrine fashion to regulate numerous cellular processes important for maintaining breast tumor growth such as pro-survival, inflammation, and invasion. S1P's and LPA's link to cancer signaling makes them tumor-promoting phospholipids with high potential for driving specific patient's disease state.[5-9]

A new assay using an impedance biosensor designed to quantify cellular adhesion changes was developed to measure phospholipid-GPCR-initiated signaling activity and the involvement of the PI3K node in transducing this activity in individual patients' live tumor cells. The CELx PI3K Signaling Function (CELx PI3K) Test measures *ex vivo* live tumor cell response in real-time to specific S1P and LPA agonists and PI3K antagonists to diagnose breast tumors with PI3K-involved hyperactive signaling.

Using primary patient breast tumor cells and well characterized breast cancer cell lines, this study set out to:

1. Characterize the involvement of the PI3K node in hyperactive S1P and LPA-initiated signaling
2. Assess whether PI3K-involved hyperactive S1P and LPA signaling is limited to breast cancer cells with PI3KCA mutations.

Methods

Breast Cancer Cell Lines: Three p110α mutant cell lines were studied. HCC1954 and BT20 were maintained according to ATCC recommendations; Cal 51 was maintained according to DSMZ recommendations. Cell lines were authenticated by ATCC STR Profiling.

Xenograft: 4-5-week-old female NSG (NOD scid gamma; NOD.Cg-*Prkdc*^{scid} Il2rg^{tm1Wjl}/SzJ) mice were injected subcutaneously in the left mammary fat pad with two million cells in 150 μL of a 50% Matrigel solution. Mice were randomly assigned to either the vehicle control group (10% Captisol; n=8) or taselisib group (10 mg/kg; n=8). After the average tumor volume reached 150 mm3, mice were dosed by oral gavage with 100 μL dosing solution daily (QD) for 28 days. Tumor size and body weight was measured at least twice weekly. Mice were euthanized at the study endpoint or when tumor size was ≥ 2 cm3 (n=2 mice euthanized prior to end of dosing; vehicle group).

Tissue Specimens: A panel of 17 fresh HER2-/PIK3CA WT tumor cells from breast cancer patients were obtained. Methods for tissue extraction and primary cell culture are essentially as described previously.[10,11] Sequences of exons 7, 9, and 20 of the PIK3CA gene were determined by PCR amplification of genomic DNA followed by Sanger sequencing utilizing exon-specific assays developed and performed by Genewiz.

CELx Test: Real-time live cell responses to an S1P agonist, an alpha-specific PI3K antagonist (alpelisib), a gamma-specific PI3K antagonist (IPI-549), and a pan-PI3K inhibitor (taselisib) were measured and quantified using an xCELLigence RTCA impedance biosensor (ACEA Biosciences). From these responses, S1P-initiated signaling, and the net amount of PI3K isoform participation in S1P-initiated signaling was quantified. A previously determined cutoff was used to identify cell samples with abnormal levels of PI3K-involved signaling activity.

Figure 1: Biosensor Enables Quantification of Signaling Activity Real-Time in Live Cells

Background

- The CELx Test leverages connections between cell adhesion, impedance, and cell signal transduction
- Live cells are attached via ECM to a microelectrode on the bottom of a 96-well impedance biosensor plate. Additionally, the tumor epithelial cells in the wells form adhesion-based gap junctions.
- The cells attached to the biosensor impede the flow of electrons when mV AC current is applied and changes in impedance (mΩ) are recorded
- Signaling activity causes cell adhesion changes that affect impedance levels recorded by the biosensor

Example Impedance vs. Time Data Set

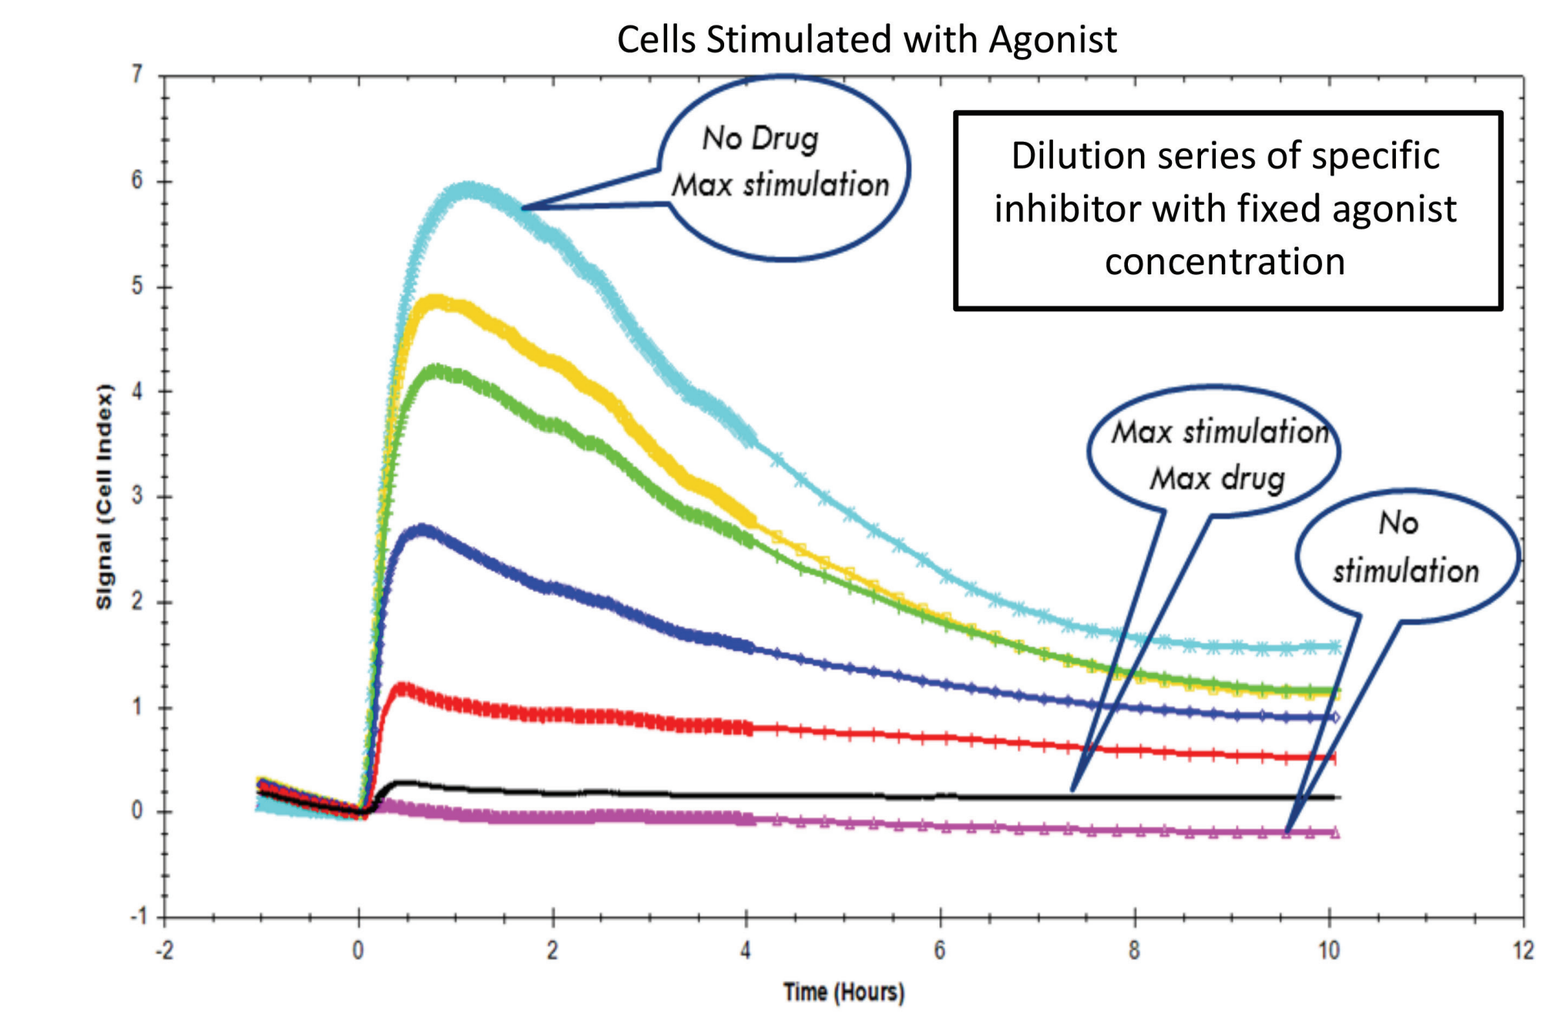
Cells Stimulated with Agonist
No Drug
Max stimulation
Dilution series of specific inhibitor with fixed agonist concentration
Max stimulation
Max drug
No stimulation
Signal (Cell Index)
Time (Hours)
7
6
5
4
3
2
1
0
-1
-2
0
2
4
6
8
10
12

Pathway specific ligands and inhibitors used to turn on/off signaling pathways

Very subtle cell adhesion changes are measured

Response quantified as signaling units over time to characterize signaling activity

Determine Signaling Score for each pathway tested

Results

Figure 2: Analysis of PI3K Involvement in S1P/LPA/HER1/HER3 Signaling in p110α Mutant HCC1954

- Two PI3K-isoform inhibitors and one pan-PI3K inhibitor were tested using CELx to quantify potential PI3K isoform activity associated with S1P, LPA, HER1, & HER3 agonism.
- CELx analysis found that activity involving the PI3K-α isoform is low and normal in all three agonist activities, despite presence of p110α mutations
- Overall pan-PI3K activity involving S1P and LPA when determined using taselisib (in green) is reported as abnormal when above test cut-off

	P13K-pan (Taselisib)	P13K-α (Alpelisib)	P13K-γ (IPI-549)
S1P (S1P)	668	196	0
LPA1 (LPA1)	1151	143	0
HER1 (EGF)	163	0	0
HER3 (NRG1)	0	0	0

- Data is reported in Signaling Units, which represents the signaling activity associated with a pathway and the binding target of the inhibitor
- S1P, LPA, EGF, NRG1 individually at 100 nM, 1000 nM, 300 pM, 50 pM respectively
- Taselisib, alpelisib, and IPI-549 individually at 333 nM

Figure 3: HCC1954 Xenograft Response to pan-PI3K Inhibitor, Taselisib

- **Taselisib induces a significant anti-tumor effect in the HCC1954 tumor**
 - T/C ratio = 0.21 (p = 0.009; t-test)
- These results are consistent with the Taselisib CELx Test data that reported abnormal levels of signaling activity involving pan-PI3K isoforms initiated by S1P or LPA activation
 - **Provides strong evidence that abnormal S1P or LPA signaling through the PI3K node is oncogenic**
- A previously reported xenograft study found alpelisib, a PI3K-α inhibitor, had no anti-tumor effect on HCC1954.[12]
 - This data is consistent with the CELx Test data reporting that S1P and LPA signaling involving PI3K-α is at normal levels
 - Provides strong evidence that p110α mutants tumors lacking abnormal PI3K involved S1P or LPA signaling are not likely to benefit from a PI3K-α specific inhibitor

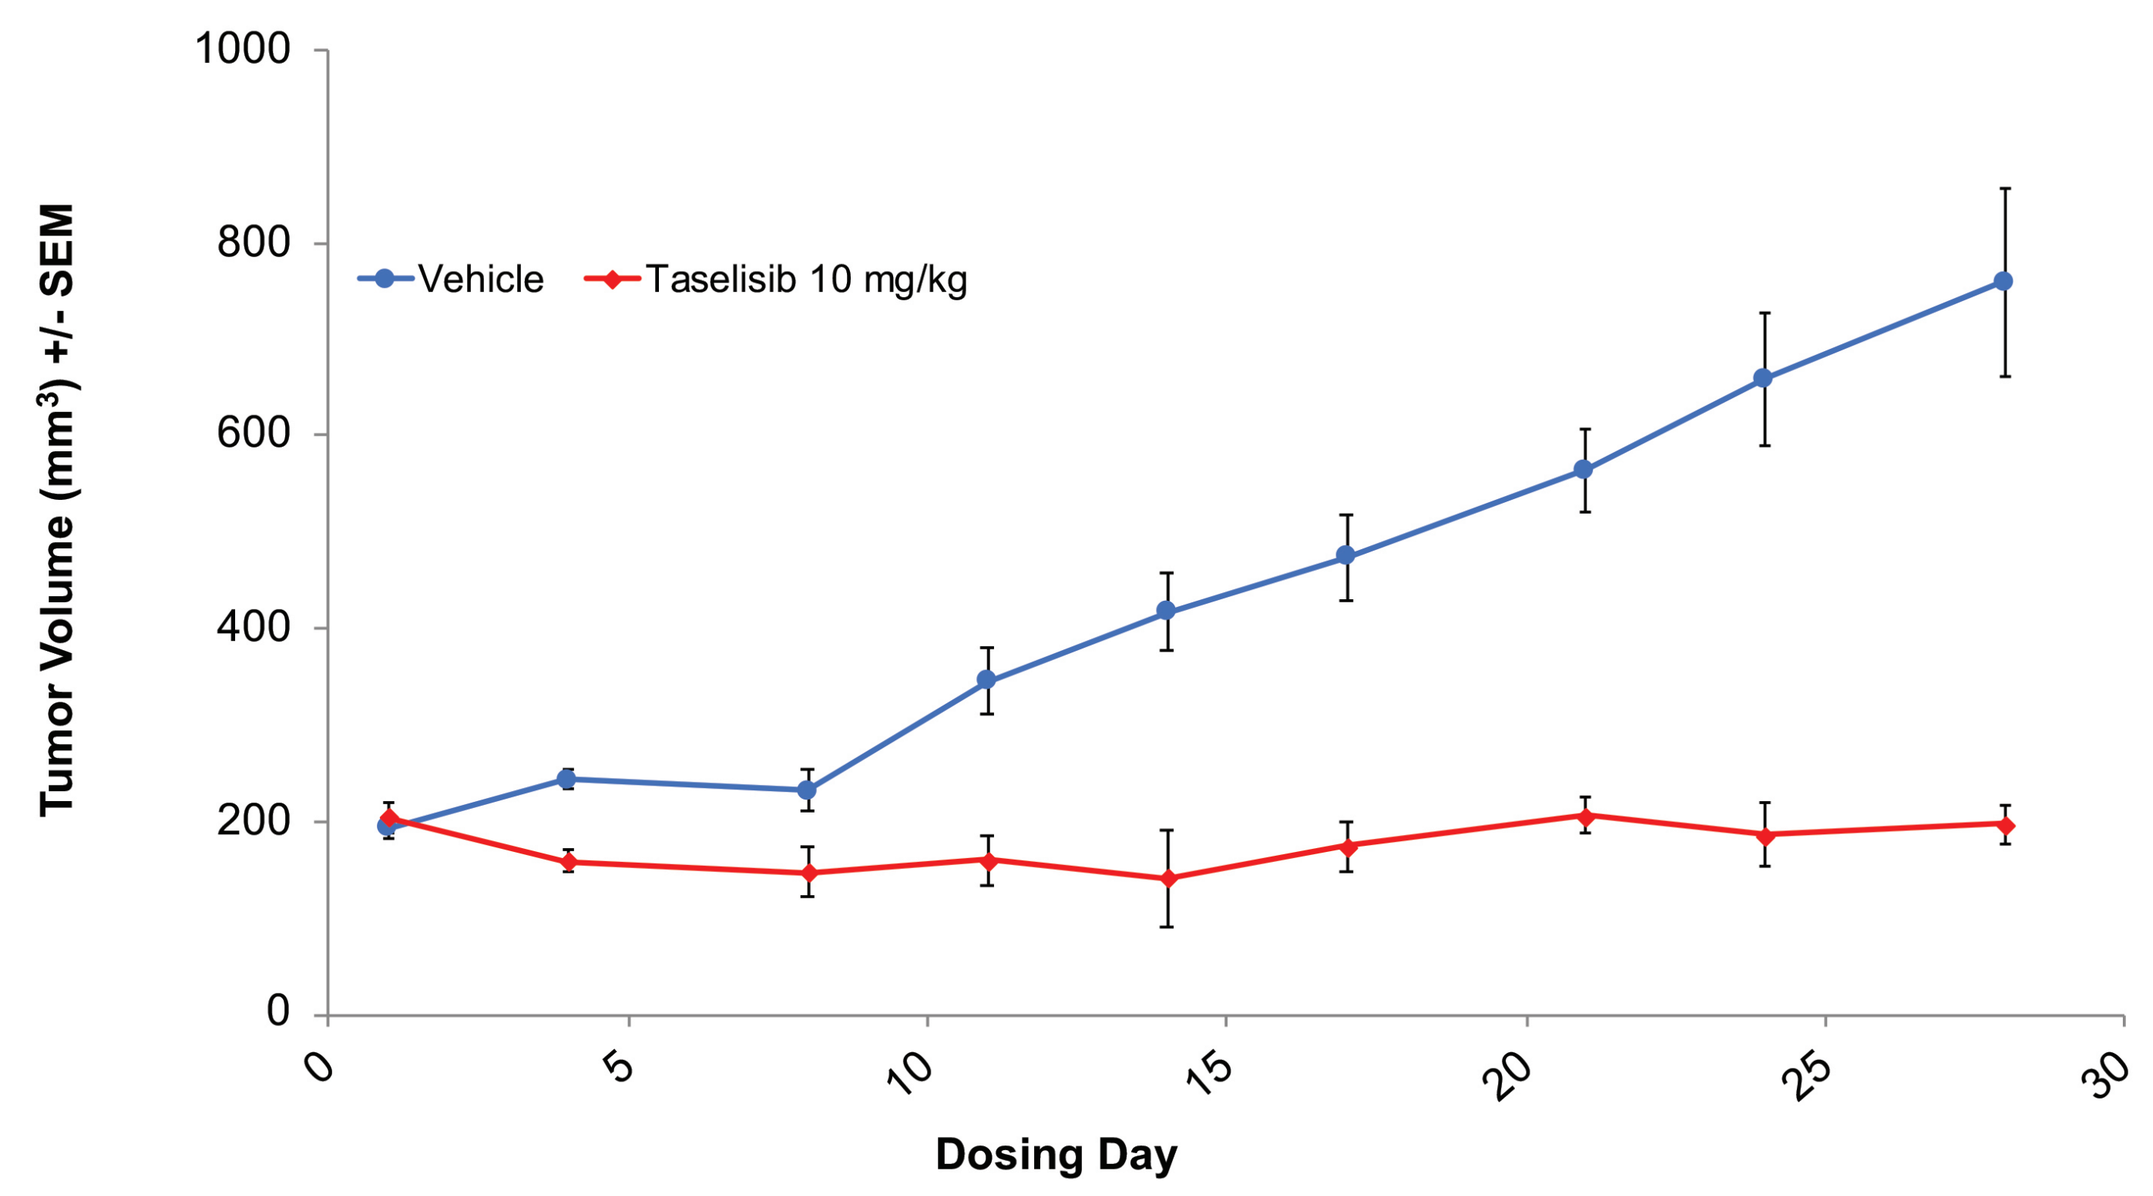
Vehicle
Taselisib 10 mg/kg
Tumor Volume (mm³) +/- SEM
Dosing Day
1000
800
600
400
200
0
0
5
10
15
20
25
30

Figure 4: Analysis of PI3K Involvement in S1P & LPA Signaling in Three p110α Mutant Cell Lines

- Two PI3K-isoform inhibitors and one pan-PI3K inhibitor were tested in CELx to quantify potential PI3K isoform activity associated with S1P or LPA agonism in additional mutant cell lines
- Abnormal activity involving PI3K-α was only found in 1 of 3 the cell lines tested (in yellow)
 - No cell lines had abnormal PI3K-γ activity
- Two cell lines had abnormal S1P and LPA activity involving pan-PI3K-isoforms (in green)
- A previously reported xenograft study found alpelisib had no anti-tumor effect on CAL-51.[12]
 - Provides further evidence that if S1P or LPA activity involving PI3K-α is normal, a PI3K-α inhibitor is not likely to provide benefit

	SIP Activated		LPA Activated	
Cell Line	Pan-P13K (Taselisib)	P13K-α (Alpelisib)	Pan-P13K (Taselisib)	P13K-α (Alpelisib)
BT20	838	331	1496	517
HCC1954	668	196	1151	143
CAL-51	189	103	147	0

- Data is reported in Signaling Units, which represents the signaling activity associated with a pathway and the binding target of the inhibitor
- S1P tested at 100 nM, LPA at 500 nM, taselisib, alpelisib at 333 nM

Results

Figure 5: Abnormal PI3K Signaling Detected in PIK3CA WT Patient Population

	S1P activated PI3K Signaling Activity			LPA activated PI3K Signaling Activity		
Patient Cell Sample	PI3K-α (Alpelisib)	PI3K-γ (IPI-549)	PI3K-α + PI3K-γ Total	PI3K-α (Alpelisib)	PI3K-γ (IPI-549)	PI3K-α + PI3K-γ Total
C270	112	203	314	197	343	540
C223	66	208	274	26	206	233
C230	109	193	302	60	360	420
C152	155	215	370	85	476	562

- Data is reported in Signaling Units, which represents the signaling activity associated with a pathway and the binding target of the inhibitor
- S1P and LPA individually at 100 nM and 500 nM respectively; alpelisib and IPI-549 individually at 333 nM

- 17 HER2-negative breast cancer cell samples obtained from patient tumors were evaluated using the CELx Test
 - Each patient cell sample was confirmed to have no p110-alpha mutations or variants using Sanger sequencing analysis of the p110-alpha gene
- A CELx PI3K Test was performed on each sample using two PI3K-isoform inhibitors to quantify potential PI3K isoform activity associated with S1P and LPA agonism.
- Of the 17 patient cell samples tested, four were found to have total levels of signaling activity involving PI3K isoforms initiated by S1P activation above the 250 signaling unit test cut-off.
 - Confirms that abnormal S1P or LPA activity involving PI3K can occur in PIK3CA WT patient primary cells.

Summary of Results

- CELx PI3K Test results correlate with xenograft results using two different cell lines with p110α-mutations.
 - Abnormal pan-PI3K signaling initiated by GPCR's found in the HCC1954 cell line corresponded with the significant anti-tumor effect found in the pan-PI3K inhibitor (taselisib) arm.
 - Normal PI3K-α isoform involved signaling coincided with previously reported results finding no anti-tumor effect with a PI3K-α isoform inhibitor (alpelisib) in HCC1954 and CAL-51.[12]
- The CELx PI3K Test found two of three cell lines with p110α-mutations lacked abnormal PI3K-α isoform involved signaling.
- A fraction of p110α-WT patient tumor samples have abnormal PI3K involved signaling.

Conclusions

- We have previously reported that: i) hyperactive HER2 signaling and hyperactive HER/c-Met signaling occurs in a sub-group of HER2-/c-Met- breast cancer patients, and ii) xenograft models demonstrate that using corresponding TKI's to inhibit this hyperactive signaling induces significant anti-tumor effect.[10,13,14]
- We now extend those studies to demonstrate how receptors from different classes (RTK and GPCR) work in unexpected ways to regulate complex signaling through an internal kinase to control various cellular functions associated with PI3K isoforms, independent of the mutational status of p110α.
- Measurement of PI3K-involved signaling initiated by GPCR's may provide a more sensitive and specific method of identifying patients most likely to benefit from PI3K inhibitors than p110α status.

References

1. Andre F., et al. Alpelisib for PIK3CA-Mutated, Hormone Receptor–Positive Advanced Breast Cancer. N Engl J Med 2019;380:1929-40
2. Fruman and Cantley, et al. The PI3K Pathway in Human Disease. Cell 2017. 170:605-635.
3. Hu and Cantley, et al. Phosphoinositide 3-Kinase Regulates Glycolysis through Mobilization of Aldolase from the Actin Cytoskeleton. Cell. 2016.; 164, 433–446
4. Currie et al, Cellular Fatty Acid Metabolism and Cancer. Cell Metabolism 2013; 18:, 153-161.
5. Ogretmen, B., Hannun, Y. Biologically active sphingolipids in cancer pathogenesis and treatment. *Nat Rev Cancer* 2004; 4, 604–616
6. Ohotski J, et al. Identification of novel functional and spatial associations between SK1, S1P and other signaling proteins that affect prognostic outcome in ER-positive breast cancer. Int J Cancer. 2013. 132(3):605-16
7. Pyne NJ et al. Sphingosine 1-phosphate receptors and sphingosine kinase 1: novel biomarkers for clinical prognosis in breast, prostate, and hematological cancers. Front Oncol. 2012 2: 168.
8. Valdés-Rives and González-Arenas Autotaxin-Lysophosphatidic Acid: From Inflammation to Cancer Development. Mediators of Inflammation. 2017. 9173090
9. Prestwich G, et al. Dual Activity Lysophosphatidic Acid Receptor Pan-Antagonist/Autotaxin Inhibitor Reduces Breast Cancer Cell Migration In vitro and Causes Tumor Regression In vivo. Cancer Res 2009; 69 (13)
10. Laing L., et al. Development of a test that measures real-time HER2 signaling activity in live breast cancer cell lines and primary cells. BMC Cancer. 2016. 17:199.
11. Stampfer MR, et al. (2002). Culture of human mammary epithelial cells. In: Freshney RI and Freshney MG, eds. Culture of Epithelial Cells.
12. Elkabets M. et al. mTORC1 Inhibition Is Required for Sensitivity to PI3K p110a Inhibitors in PIK3CA-Mutant Breast Cancer. Sci Transl Med 5, 196ra99 (2013);
13. Laing L., et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity. Oncotarget. 2016. 7(48):78577-78590.
14. Laing et al., Evaluation of pan-HER and c-MET inhibitors tested in primary HER2- breast cancer cells with hyperactive c-MET and ErbB family signaling, Cancer Research;79 (4 Supplement) P2-05-05

Abstract #: P1-09-07

This presentation is the intellectual property of Celcuity Inc. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.